A NEW QUANTITATIVE METHOD FOR THE MEASUREMENT OF GROSS MORPHOLOGICAL CARTILAGE CHANGES IN ANIMAL MODELS OF OSTEOARTHRITIS.

Karl A. Rudolphi*, **Michael Grunkin****, **Johan Doré****

*Aventis Pharma Deutschland GmbH, Frankfurt/Main, Germany; **Visiopharm ApS, Hørsholm, Denmark

Methods

Staining Procedure: 120 male STR/1N mice, aged between 6 and 21 weeks, were sacrificed by CO_2 inhalation. Their right hindlegs were removed and the tibiae were carefully isolated, freed of adjacent soft tissue without damaging the knee joint surface and stored in a moist chamber until stained as follows: the tibiae were held with a forceps at the distal stump, immersed in a solution of 0.1% Toluidine-Blue for 30 sec and rinsed with 5 ml of saline (0.9%). The stained tibiae were then air dried at room temperature and stored in Eppendorff vials until image aquisition.

Image acquisition and -analysis: For image acquisition the specimen were positioned vertically in a holding device with the stained tibial plateaus pointing upwards, illuminated with a diffuse ring-light and finally submerged in water in order to avoid reflections on the joint surface(Figure 3). A Leica Wild M651 microscope with a CCD camera Leica DC 300 with 1000 pixels per mm image resolution was used to acquire high-resolution color digital images of the medial side of the knee joint. A calibration method for microscopy was developed, allowing for the possibility that measurements of joint degradation may be compared directly across studies and time. The OAIx/S method has been implemented and integrated into the Windows based VIS system developed by Visiopharm (Figure 4). This system enables the operator to acquire images and store them in a database where they can be integrated with all other study data. Measurements can be obtained fast and efficiently through a simple and intuitive user interface. The results and statistical analysis are displayed graphically and the results are easily exported to other standard software packages.

Aim of the Study

Gross morphological changes of the articular cartilage such as fibrillations up to complete erosions are hallmarks of joint pathology in osteoarthritis (OA). In experimental animals these changes are mostly evaluated by subjective semi-quantitative scoring after marking the cartilage erosions with India-ink. Unfortunately, this staining procedure is not applicable to small experimental animal species such as mice, rats and guinea pigs, because the ink particles are to big to mark the cartilage lesions in the small joints. Based on digital image analysis, a novel technique (the joint degradation index, OAIx/S) was developed to visualize and quantify the cartilage changes in the knee joints of STR/1N mice - a strain which develops moderate to severe OA spontaneously during the first 6 months of their life (Figure 2).

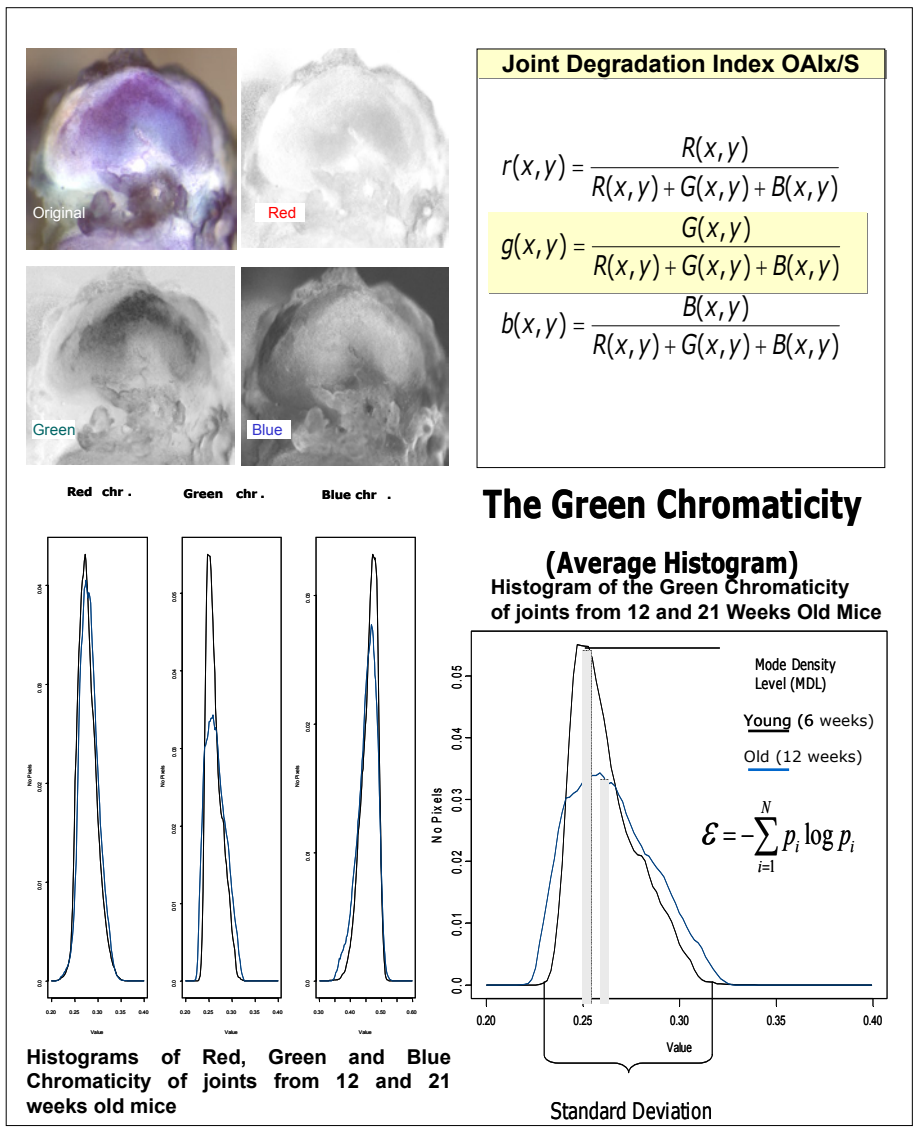
Original
Red
Green
Blue
Joint Degradation Index OAIx/S
$r(x,y) = \frac{R(x,y)}{R(x,y)+G(x,y)+B(x,y)}$
$g(x,y) = \frac{G(x,y)}{R(x,y)+G(x,y)+B(x,y)}$
$b(x,y) = \frac{B(x,y)}{R(x,y)+G(x,y)+B(x,y)}$
Red chr.
Green chr.
Blue chr.
Histograms of Red, Green and Blue Chromaticity of joints from 12 and 21 weeks old mice
The Green Chromaticity
(Average Histogram)
Histogram of the Green Chromaticity of joints from 12 and 21 Weeks Old Mice
Mode Density Level (MDL)
Young (6 weeks)
Old (12 weeks)
$\mathcal{E} = -\sum_{i=1}^{N} p_i \log p_i$
No Pixels
Value
Standard Deviation

Figure 1: Theoretical Basis of the OAIx/S Joint Degradation Index - The Tri-Chromatic Color Model -

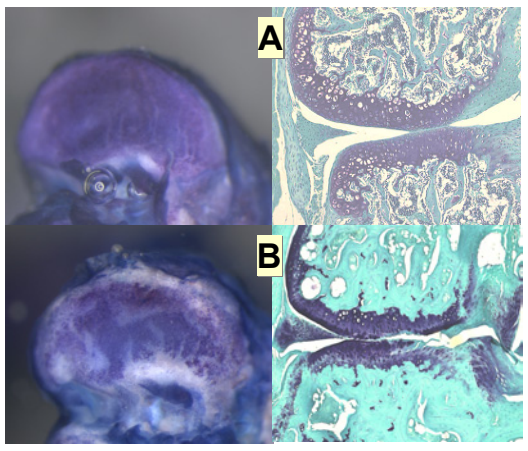
A
B

Figure 2:Macroscopic (left, tibial plateau) and microscopic (right, sagittal section of complete joint) images of Toluidine Blue – stained knee joints of STR/1N mice without (A) and with (B) osteoarthritic destruction of the cartilage

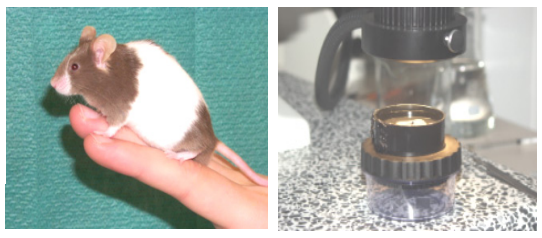

Figure 3: STR/1N Mouse and Mouse Tibia Positioned Under the Microscope

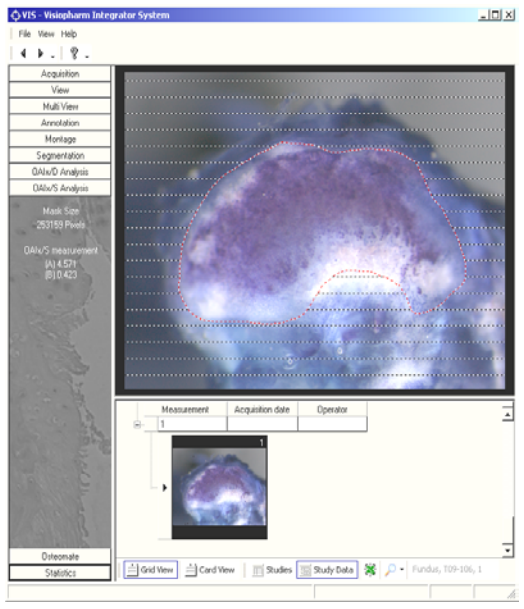
VIS - Visiopharm Integrator System
Acquisition
View
Multi View
Annotation
Montage
Segmentation
OAIx/D Analysis
OAIx/S Analysis
Measurement
Acquisition date
Operator
Osteomate
Statistics

Figure 4: Screendump of the *VIS* User Surface Showing a Medial Tibial Plateau With Marked Region of Interest (ROI)

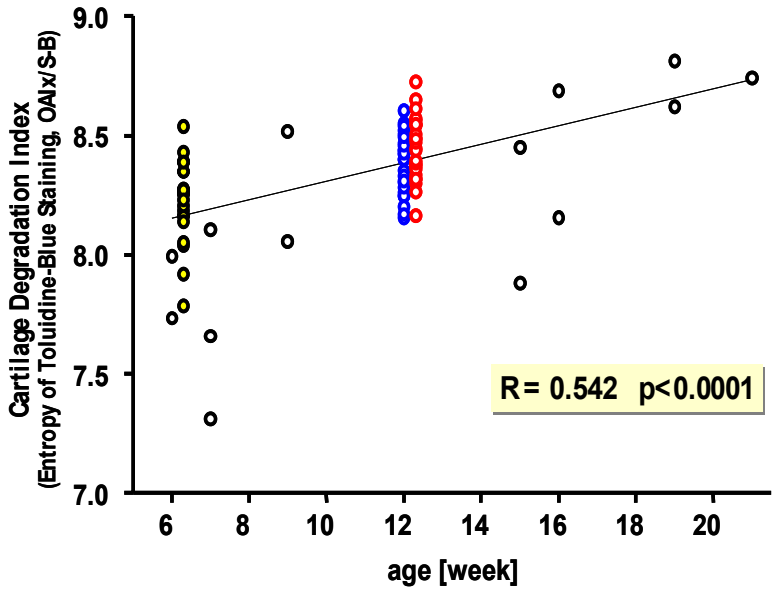
Cartilage Degradation Index
(Entropy of Toluidine-Blue Staining, OAIx/S-B)
9.0
8.5
8.0
7.5
7.0
6
8
10
12
14
16
18
20
age [week]
R= 0.542 p<0.0001

Figure 5: Progessive Aggravation of Joint Damage in STR/1N Mice as
Demonstrated by a Significant Correlation (Linear Regression Analysis) of the Entropy of Toluidine-Blue Staining of the Medial Tibial Plateau to Increasing Age.

Conclusion

The results found in this age-dependency study are very promising, and warrant further validation in larger studies including additional parameters such as biomarkers of joint destruction and histopathology. If consistent results continue to validate the method, it may be useful as a primary decision parameter. This would potentially reduce analysis time significantly, as preparation of the specimens can be done in less than a week and the analysis of an entire study can be done in less than a day. A total histological analysis requires at least 4-6 weeks. The generality of the OAIx/S method makes it quite likely that it can be used directly, or with relatively minor modifications, on other small rodent species such rats and guinea pigs.

Results

The Toluidine Blue staining significantly enhanced the contrast between intact cartilage and OA lesions (Figures 1 and 2).

The testing of a number of features in different color representations shows that features which describe the spread of the histogram for the trichromatic green channel provides the best description of cartilage degradation (Figure 1).

The OAIx/S method was generally found to be quite robust towards minor variations in staining intensity and illumination. By using the entropy as a scale-independent measure of joint degradation, it is possible to detect the age-dependent aggravation of osteoarthritic changes. The entropy feature was found to correlate strongly with age (R = 0.54, p <0.0001) as seen in Figure 5.

Both, concerning region of interest (ROI) drawing variability and image acquisition variability, the entropy feature shows a good reproducibility with a precision error found to be in the interval of 0.6%-1.5% based on two independent measurements. This level is well below the changes to be expected with biologically meaningful effects in pharmacological studies. Ongoing experiments will show if this method is indeed suitable to detect such treatment effects of test compounds.

Addresses for Correspondence:
K. Rudolphi:
Aventis Pharma Deutschland GmbH
Industriepark Höchst, Bldg. H-821
D-65926 Frankfurt/Main, Germany
☺ ++49-69-305-15299
Karl.Rudolphi@Aventis.com
M. Grunkin & J. Doré:
Visiopharm
Bøge Allé 5
DK-2970 Hørsholm
Denmark
☺++45-7240-2990
mgr@visiopharm.com
jdh@visiopharm.com
http://www.visiopharm.com

Acknowledgements:
We thank Kamilla Sedlak, Alexandra Pabst and Wolfgang Harangozo for the excellent technical execution of the experiments.

For excellent data analysis and software development we thank Lars Peders, Steen Rasmussen and Thomas Jensen.